Open-Label Extension of a Randomized, Double-Blind, Placebo-Controlled, Phase 3 Study of the Safety and Analgesic Efficacy of MNK-795 Controlled-Release Oxycodone/Acetaminophen Tablets (CR OC/APAP) in an Acute Pain Model

Neil Singla, MD[1]; Thomas Barrett, PhD[2]; Lisa Sisk[2]; Kenneth Kostenbader, MD[2]; Jim Young, PhD[2]; Michael Giuliani, MD[2]
[1]Lotus Clinical Research LLC, Pasadena, CA, USA; [2]Mallinckrodt Inc., Hazelwood, MO, USA

Mallinckrodt Pharmaceuticals

Introduction

- Despite the wide range of treatment options available for acute pain, advances in the management of acute pain are needed[1]
- Multimodal therapy combining oxycodone (OC) and acetaminophen (APAP) is a well-established approach to the treatment of acute pain[2,3]
 - Combining agents with different mechanisms of action may offer additive effects, while allowing for the management of pain at a lower dose of each component, potentially reducing the risk of concentration-dependent adverse events[4-6]
- In addition, formulations engineered to provide quick and sustained release may offer therapeutic benefit as well as reduce the pill burden[5,7]
- MNK-795 (CR OC/APAP) is a controlled-release (CR) combination OC/APAP analgesic, and is being designed to provide both fast onset of analgesia within 1 hour and sustained analgesia over the 12-hour dosing interval
 - CR OC/APAP tablets employ a dual-layer biphasic delivery mechanism that, when administered as a single dose (ie, 2 tablets), ensures the IR component delivers 3.75 mg OC/325 mg APAP and the extended-release component delivers 11.25 mg OC/325 mg APAP
 - Incorporates technology designed to provide tamper resistance and abuse deterrence
- In this pivotal clinical trial, CR OC/APAP was studied in an established acute pain model in patients undergoing a first metatarsal bunionectomy; medication effects were evaluated 48 hours post-procedure (double-blind) and continued throughout a voluntary open-label treatment period (up to 14 days)

Objectives

- Evaluate the safety of the administration of multiple doses of CR OC/APAP in patients with moderate to severe acute pain in an open-label extension phase of a randomized, double-blind, placebo-controlled, phase 3 study

Methods

PATIENTS

- Patients aged 18 to 75 years undergoing unilateral, first metatarsal bunionectomy who reported at least moderate or severe pain intensity and numeric rating scale score of ≥4 (out of 10) between the hours of 4:00 AM and 12:00 PM (after cessation of intravenous popliteal nerve block) on the first postoperative day were eligible for the study

STUDY DESIGN

- Multicenter, randomized, double-blind, placebo-controlled, parallel group, phase 3 study of CR OC/APAP in patients with moderate to severe acute pain
 - Screening period of between 2 and 30 days before surgery, a randomized double-blind dosing phase of 2 days (48 h) postprocedure, and an optional ≤14 day open-label extension for qualified patients (**Figure 1**)

FIGURE 1: Study Design

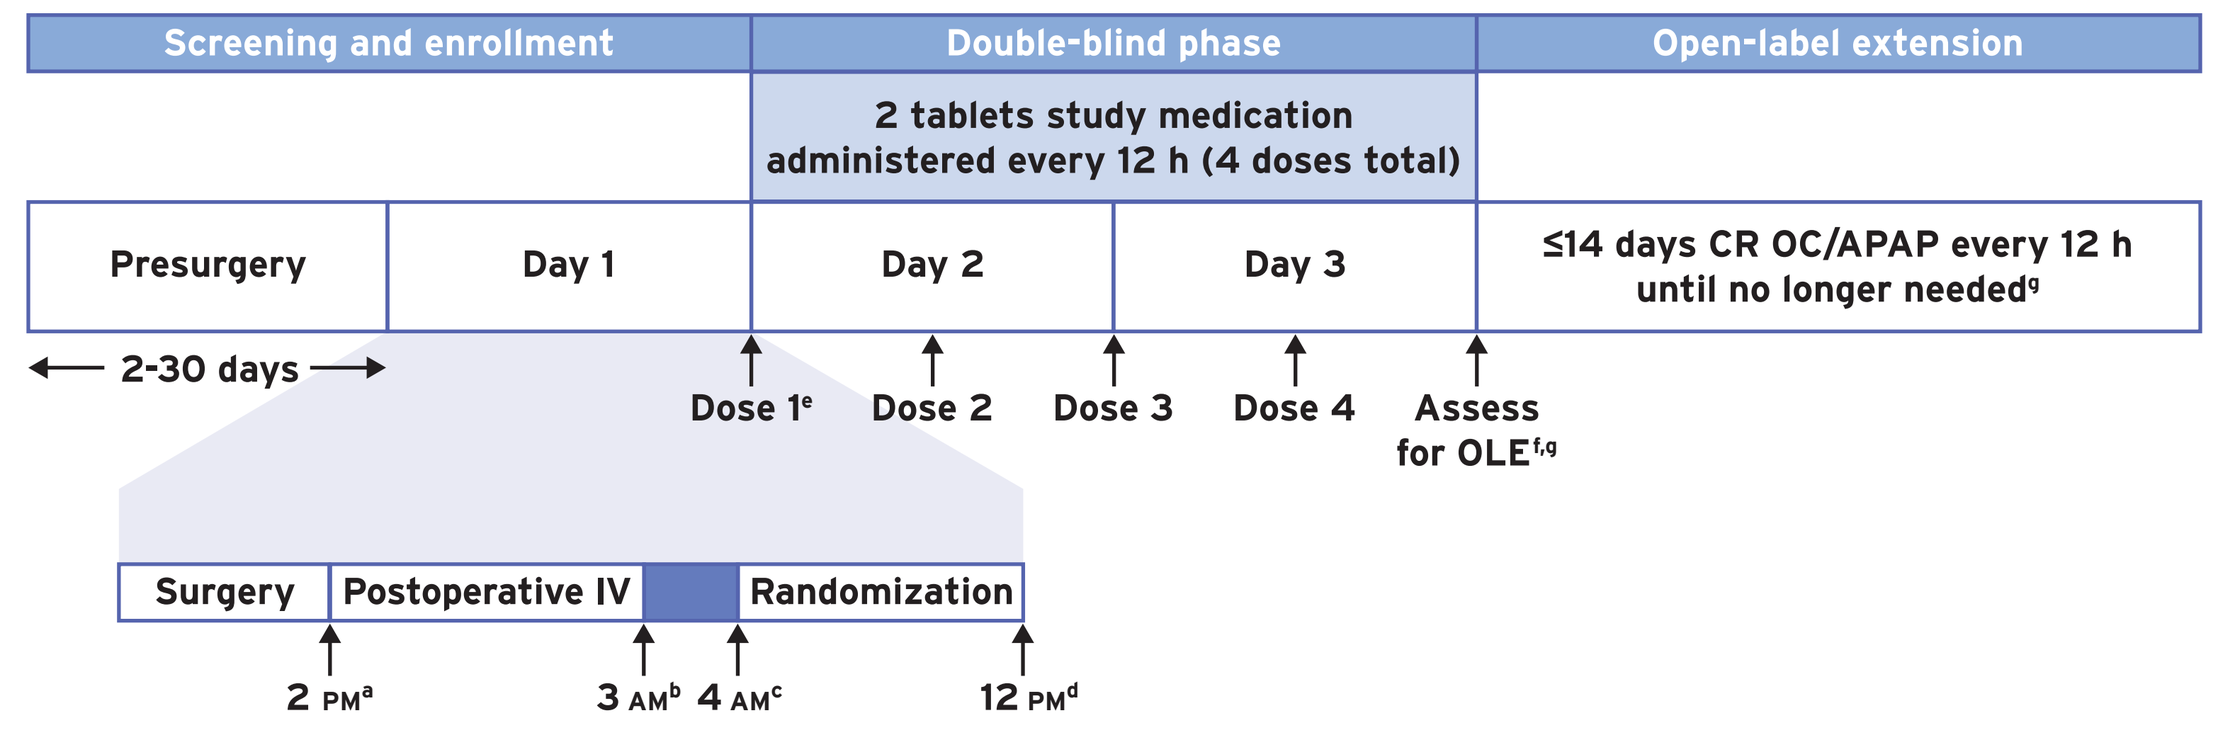
Screening and enrollment
Double-blind phase
Open-label extension
2 tablets study medication administered every 12 h (4 doses total)
Presurgery
Day 1
Day 2
Day 3
≤14 days CR OC/APAP every 12 h until no longer needed[g]
2-30 days
Dose 1[e]
Dose 2
Dose 3
Dose 4
Assess for OLE[f,g]
Surgery
Postoperative IV
Randomization
2 PM[a]
3 AM[b]
4 AM[c]
12 PM[d]

[a]Surgery completed; [b]Nerve block stopped; [c]Earliest start for pain assessment and randomization; [d]Latest start for pain assessment and randomization; [e]Study medication administered within 30 minutes of randomization; [f]Patients assessed for participation in open-label extension within 48 to 52 hours of receiving first dose of study drug; [g]End-of-treatment evaluations performed within 3 days of receiving last dose of study drug; follow-up telephone call 7±2 days after receiving last dose of study drug (double-blind and open-label phases).

- During the double-blind phase of the study, patients were randomized to receive 2 tablets of CR OC/APAP (total 15 mg OC/650 mg APAP) or placebo administered every 12 hours (0, 12, 24, and 36 h; 4 total doses)
 - Use of supplemental analgesia was permitted (ie, ibuprofen 400 mg up to 6 times per day [2400 mg/d]) during the double-blind and open-label phases of the study
- Eligibility criteria for the open-label extension phase of the study included completing the double-blind phase of the study; having a pain intensity score ≥3 at completion of the double-blind phase of the study, but no later than 52 hours after receiving the first dose of study drug; signing an open-label extension consent form prior to surgery; and agreeing to participate in the open-label extension phase of the study
- Patients participating in the open-label phase of the study were discharged with instructions to take 2 tablets of CR OC/APAP every 12 hours *until no longer needed*
 - The open-label phase lasted *up to* 14 days, with clinic visits at days 7 and 14 (±1 day), followed by a telephone call 7 days (±2 days) after the last dose
- Exclusion criteria included any medical condition that might decrease study compliance or alter the absorption, distribution, metabolism, or excretion of the study drug (eg, severe chronic diarrhea, chronic constipation, irritable bowel syndrome, or unexplained weight loss); gastric bypass surgery or gastric band; history of intolerance to short-term opioid use; and treatment with study drug or bunionectomy in previous 3 months

ASSESSMENTS DURING THE OPEN-LABEL EXTENSION

- Safety and tolerability assessments were conducted throughout the open-label phase of the study and included physical examinations, measurement of vital signs (eg, sitting blood pressure, pulse rate, and temperature), and clinical laboratory tests (ie, chemistry, hematology, and urinalysis)
 - Adverse events were collected at each visit and the 7-day follow-up phone call
- Global assessment of patient satisfaction was evaluated at 48 hours or early termination for the blinded-dosing phase and at every clinic visit for the open-label phase
 - Assessed the patient's satisfaction with treatment across 5 dimensions, such as ease of administration and level of pain relief, on a categorical scale (ie, very satisfied, satisfied, neither satisfied nor dissatisfied, dissatisfied, or very dissatisfied)

STATISTICAL ANALYSES

- Descriptive statistics were summarized for baseline characteristics and global assessment of satisfaction
- Medication adherence and treatment-emergent adverse events (TEAEs) were summarized using frequencies and percentages
- Summary statistics for actual values and changes from baseline were calculated for the physical examination findings, laboratory test results, vital signs, and pulse oximetry, and a shift analysis examined categorical changes from baseline to various time points

Results

PATIENT POPULATION

- A total of 329 patients were enrolled and received ≥1 dose of study drug in the blinded-dosing phase of the study
 - 166 patients received CR OC/APAP; 163 received placebo
 - 293 patients (89.1%) completed the double-blind phase of the study
 - 146 patients (49.8%; prior CR OC/APAP, n=77; prior placebo, n=69) who completed the double-blind phase of the study entered the open-label phase of the study, with 129 patients (88.4%) completing the open-label extension
 - 145 patients attended the 1-week follow-up visit, and 36 patients attended the 2-week follow-up visit
- Demographic characteristics of the open-label safety population were generally similar between groups (**Table 1**)
- During the open-label dosing phase, 120 patients (82.2%) received ±20% of the expected doses

TABLE 1: Demographic and Baseline Characteristics, Open-Label Safety Population

Parameter	Prior Double-Blind CR OC/APAP (n=77)	Prior Double-Blind Placebo (n=69)	All Patients in Open-Label Phase (N=146)
Age, y, mean (SD)	39.9 (12.4)	41.4 (14.1)	40.6 (13.2)
Female sex, n (%)	67 (87.0)	55 (79.7)	122 (83.6)
Race, n (%)			
White	42 (54.5)	48 (69.6)	90 (61.6)
Black	31 (40.3)	19 (27.5)	50 (34.2)
Asian	4 (5.2)	1 (1.4)	5 (3.4)
Native Hawaiian or other Pacific Islander	0	1 (1.4)	1 (0.7)
Weight, kg, mean (SD)	71.5 (13.1)	73.2 (12.6)	72.3 (12.8)
Body mass index, kg/m^2, mean (SD)	26.0 (4.1)	26.6 (3.4)	26.3 (3.8)

SAFETY AND TOLERABILITY

- During the open-label phase, 64 patients (43.8%) experienced ≥1 TEAE (**Table 2**)
 - The most frequently reported TEAEs were primarily gastrointestinal related (nausea, vomiting, constipation) and central nervous system-related (somnolence, headache, dizziness)

TABLE 2: Treatment-Emergent Adverse Events Occurring During the Open-Label Phase

Treatment-Emergent Adverse Event, n (%)	Prior Double-Blind CR OC/APAP (n=77)	Prior Double-Blind Placebo (n=69)	All Patients (N=146)
Any TEAE	25 (32.5)	39 (56.5)	64 (43.8)
Nausea	8 (10.4)	18 (26.1)	26 (17.8)
Vomiting	3 (3.9)	8 (11.6)	11 (7.5)
Constipation	4 (5.2)	5 (7.2)	9 (6.2)
Somnolence	1 (1.3)	6 (8.7)	7 (4.8)
Headache	4 (5.2)	2 (2.9)	6 (4.1)
Dizziness	2 (2.6)	4 (5.8)	6 (4.1)
Peripheral edema	3 (3.9)	1 (1.4)	4 (2.7)
Pruritus	1 (1.3)	3 (4.3)	4 (2.7)
Infection	1 (1.3)	3 (4.3)	4 (2.7)

- 1 patient reported 3 severe TEAEs, and 1 patient reported a serious adverse event (ie, deep vein thrombosis determined by the investigator to be unrelated to treatment with the study drug)
- Changes from baseline in laboratory values (ie, hematology, serum chemistry, and urinalysis) were generally small and were similar between treatment groups during double-blind periods and similar between the double-blind and open-label periods
 - Six patients (4.1%) had alanine aminotransferase and/or aspartate aminotransferase ≥3 times the upper limit of normal values at least once during the study
 - Total bilirubin remained within the normal reference range in all 6 cases
 - None met Hy's Law criteria

- Values and changes in vital signs after 7 days of open-label treatment are shown in **Table 3**
 - Vital signs during the open-label phase were normal in >90% of patients at any visit
 - During the open-label phase, ≤1.4% of patients had shifts from normal to abnormal oxygen saturation at any time point

TABLE 3: Vital Sign Measures and Changes From Baseline After 7 Days[a] of Open-Label Treatment

Vital Sign	Baseline Value (n=146)	OL Visit Day 7 (n=145)	Change From Baseline
Systolic blood pressure, mm Hg			
Mean (SD)	117.3 (14.12)	120.6 (14.4)	3.4 (12.34)
Median	116.0	120.0	3.0
Diastolic blood pressure, mm Hg			
Mean (SD)	73.2 (9.31)	75.2 (9.07)	2.1 (8.45)
Median	72.0	74.0	2.0
Heart rate, beats/min			
Mean (SD)	73.9 (10.81)	74.1 (11.62)	0.2 (10.99)
Median	74.0	74.0	-1.0
Respiratory rate, breaths/min			
Mean (SD)	16.3 (1.94)	15.8 (1.68)	-0.5 (2.52)
Median	16.0	16.0	0.0
Body temperature, °C			
Mean (SD)	36.66 (0.50)	36.40 (0.54)	-0.26 (0.58)
Median	36.70	36.50	-0.20
Oxygen saturation, %			
Mean (SD)	97.6 (1.69)	98.0 (1.60)	0.4 (1.76)
Median	98.0	98.0	0.0

[a]By Day 14, very few patients required pain medications (n=36); therefore, data from n=145 completing day 7 are shown here.

PATIENT SATISFACTION

- At the 7-day follow-up, more than 88% (of 144 patients) indicated they were "very satisfied" or "satisfied" on all measures (**Figure 2**)
- At the 14-day follow-up, more than 83% (of 36 patients) indicated they were "very satisfied" or "satisfied" on all measures (**Figure 2**)

FIGURE 2: Proportion of Patients "Satisfied" or "Very Satisfied" With CR OC/APAP After 7 or 14 Days of Open-Label Phase Treatment

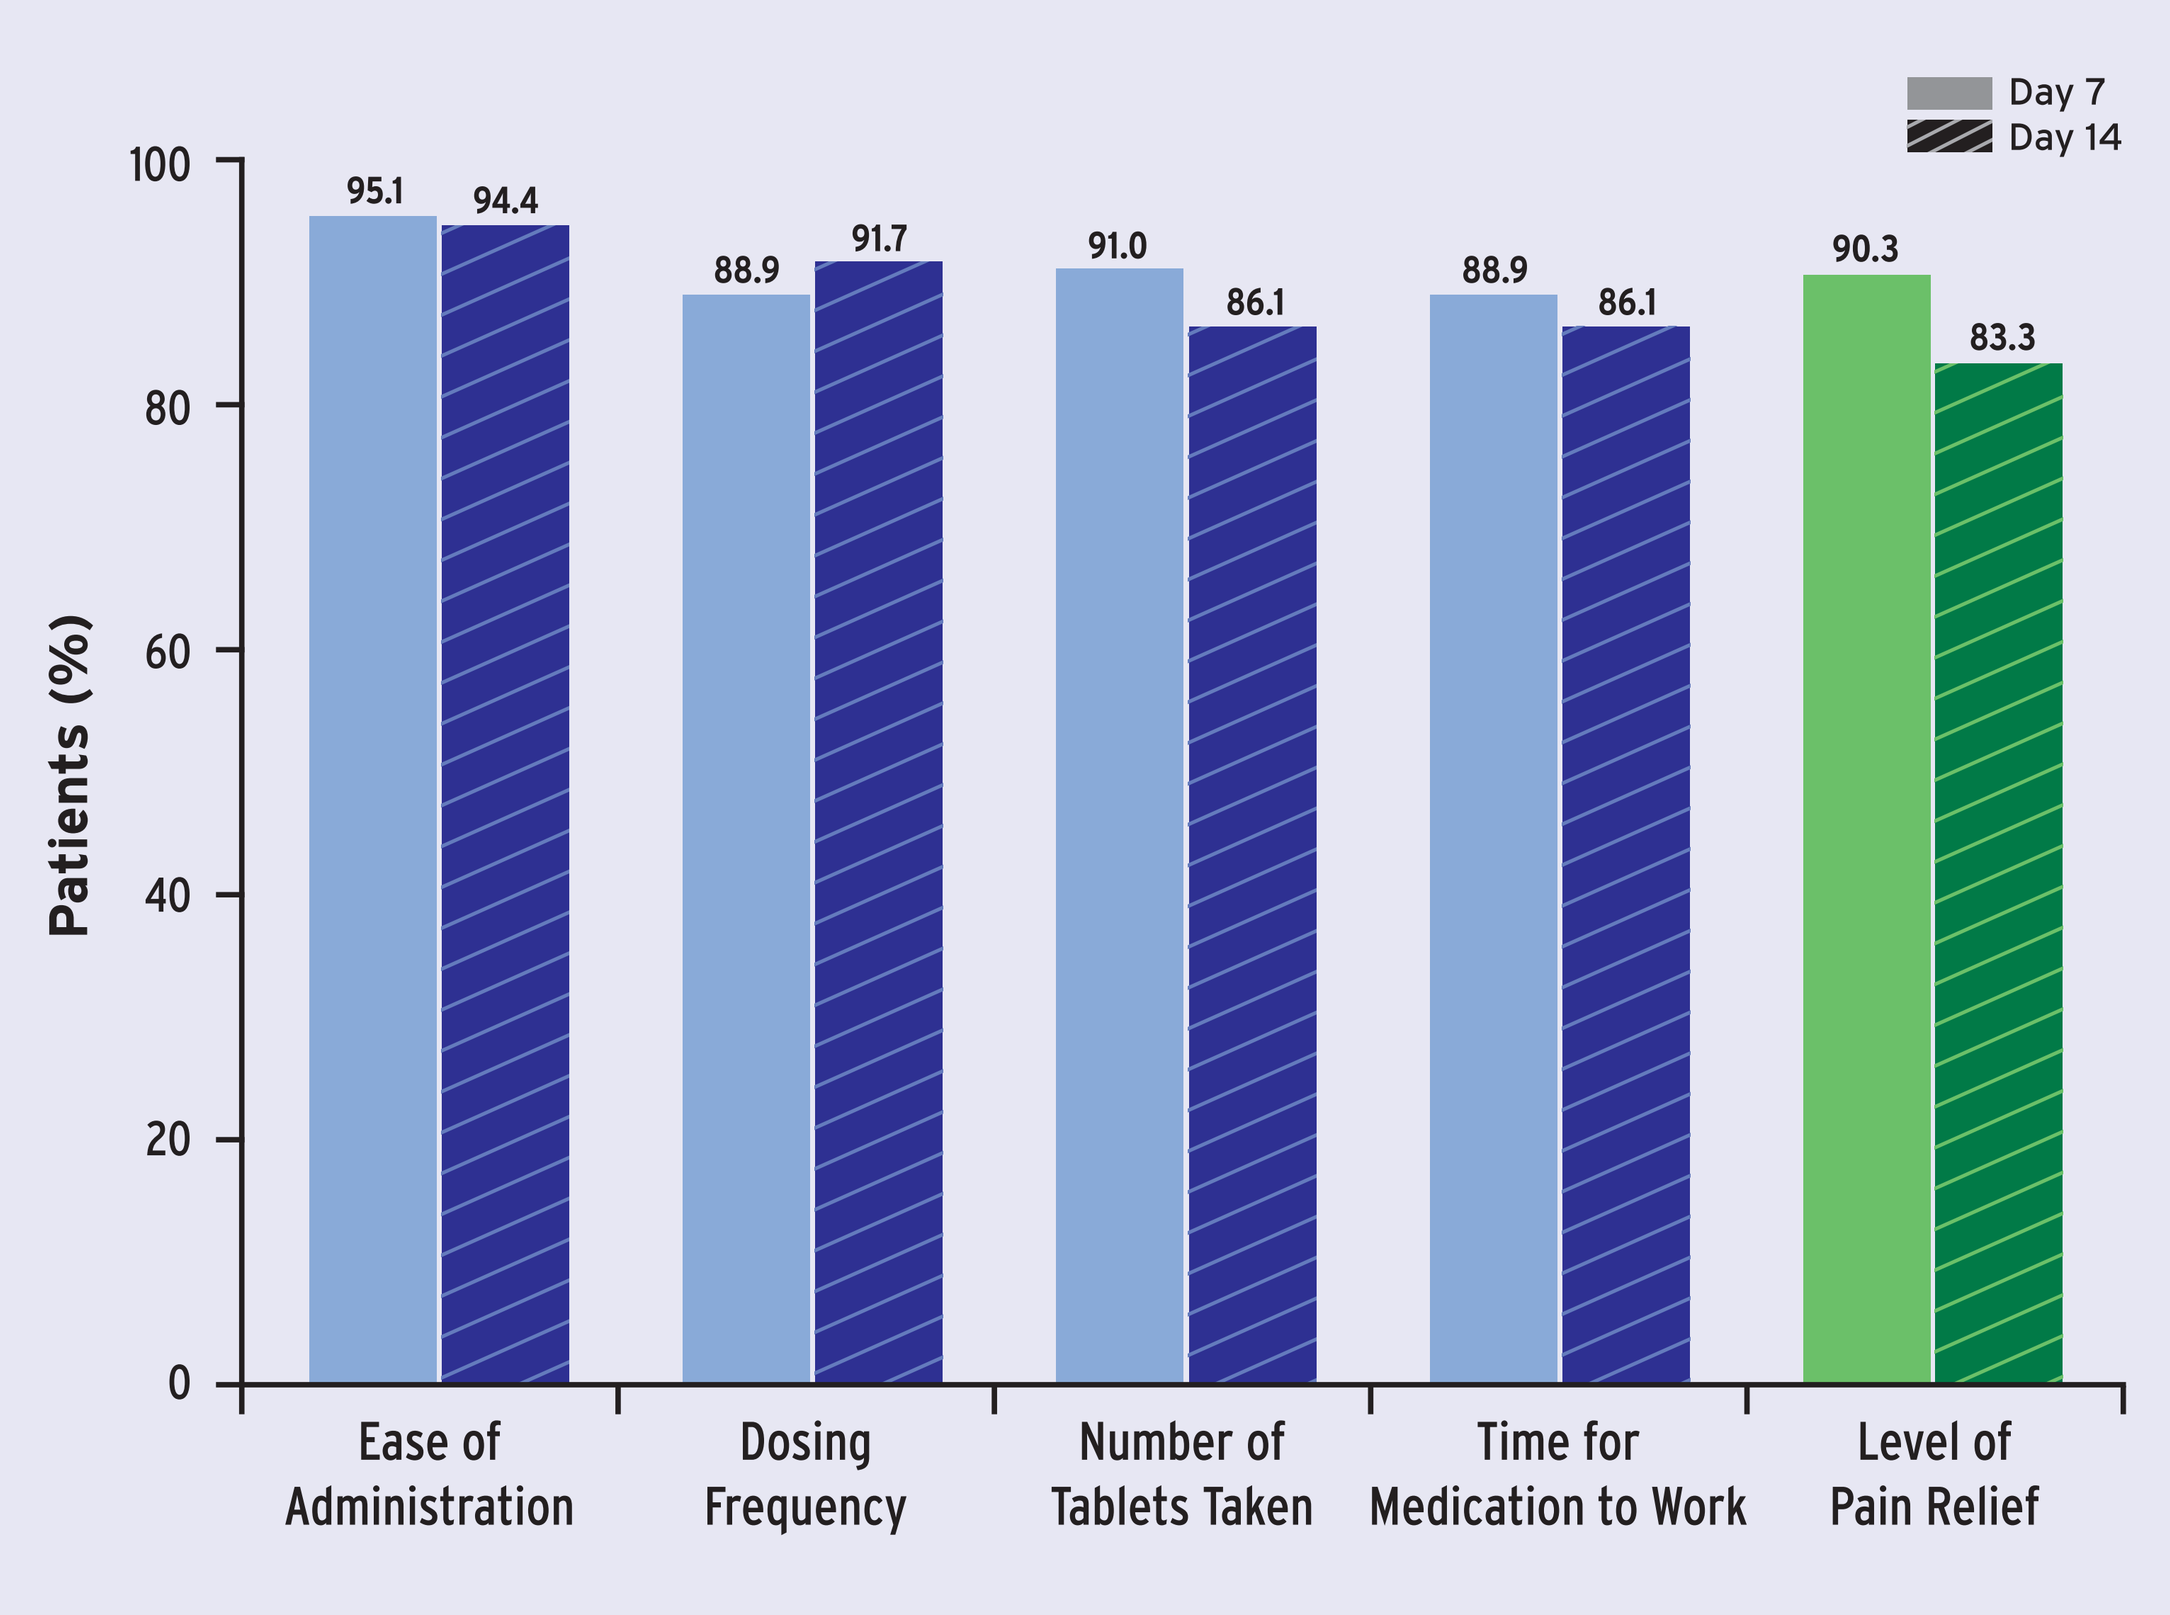
Day 7
Day 14
Patients (%)
0
20
40
60
80
100
95.1
94.4
88.9
91.7
91.0
86.1
88.9
86.1
90.3
83.3
Ease of Administration
Dosing Frequency
Number of Tablets Taken
Time for Medication to Work
Level of Pain Relief

Conclusions

- Multiple-dose administration of CR OC/APAP was generally well tolerated in this 14 day open-label extension study
 - The most frequently reported adverse events were consistent with those seen with other opioids in general, and specifically, oxycodone
 - Shifts in laboratory test results, vital signs, and oxygen saturation were generally small and not clinically significant
 - All changes in clinical laboratory tests and vital signs that were outside of the defined reference range(s) were not clinically significant according to the investigator
- More than 80% of patients were very satisfied or satisfied with every measure of treatment assessed, including 94.4% for ease of administration, 86.1% for time for the medication to work, and 83.3% for level of pain relief
- CR OC/APAP is an important addition to the armamentarium for patients with moderate to severe acute pain

References

1. Apfelbaum JL, Chen C, Mehta SS, Gan TJ. Postoperative pain experience: results from a national survey suggest postoperative pain continues to be undermanaged. *Anesth Analg*. 2003;97(2):534-540.
2. Gammaitoni AR, Galer BS, Lacouture P, et al. Effectiveness and safety of new oxycodone/acetaminophen formulations with reduced acetaminophen for the treatment of low back pain. *Pain Med.* 2003;4(1):21-30.
3. Cooper SA, Precheur H, Rauch D, et al. Evaluation of oxycodone and acetaminophen in treatment of postoperative dental pain. *Oral Surg Oral Med Oral Pathol.* 1980;50(6):496-501.
4. Barkin RL. Acetaminophen, aspirin, or ibuprofen in combination analgesic products. *Am J Ther.* 2001;8(6):433-442.
5. Raffa RB. Pharmacology of oral combination analgesics: rational therapy for pain. *J Clin Pharm Ther.* 2001;26(4):257-264.
6. Beaver WT, McMillan D. Methodological considerations in the evaluation of analgesic combinations: acetaminophen (paracetamol) and hydrocodone in postpartum pain. *Br J Clin Pharmacol.* 1980;10(Suppl 2):215S-223S.
7. McCarberg BH, Barkin RL. Long-acting opioids for chronic pain: pharmacotherapeutic opportunities to enhance compliance, quality of life, and analgesia. *Am J Ther.* 2001;8(3):181-186.

Disclosures

Dr. Singla received grants as a clinical investigator from Mallinckrodt Inc. Dr. Barrett, Ms. Sisk, Dr. Young, and Dr. Giuliani are employees of Mallinckrodt Inc. Dr. Kostenbader is a paid consultant to Mallinckrodt Inc.

Acknowledgment

Technical editorial and medical writing support for the development of this poster was provided by Sophie Bolick, PhD, Synchrony Medical Communications, LLC, West Chester, PA. Funding for this support was provided by Mallinckrodt Inc., Hazelwood, MO.

PAINWEEK 2013 | SEPTEMBER 4-7 | LAS VEGAS, NV Poster #104